Few randomized studies have evaluated optimal acute treatment and post-event prevention for patients presenting with ALI. In a modern, well characterized cohort of patients with PAD, including 508 ALI events, treatments were highly variable. Future studies evaluating optimal treatment approaches to ALI may lead an evidence base informing more systematic approaches to this severe event."

Presentation and Management of Acute Limb Ischemia Following Lower Extremity Revascularization: Insights from VOYAGER PAD Trial

•R. Wilson King, MD[1,2], Nicholas Govsyevev, MD[1,2], Mark Nehler, MD[1,2], Warren Capell, MD[1,3], Connie Hess, MD[1,2], Scott Berkowitz, MD[1], Rupert Bauersachs, MD[4], Sonia Anand, MD[5], Sebastian Debus, MD[6], Manesh Patel, MD[7], Lloyd Haskell, MD[8], Marc Bonaca, MD[1,2]

•[1]CPC Clinical Research, Aurora, CO; [2]University of Colorado Denver School of Medicine, Aurora, CO; [3]University of Colorado Denver - Anschutz Medical Campus, Aurora, CO; [4]Klinikum Darmstadt, Darmstadt, Hessen, Germany; [5]McMaster University, Hamilton, ON, Canada; [6]Universitatsklinikum Hamburg Eppendorf Universitares Herzzentrum Hamburg GmbH, Hamburg, Germany; [7]Duke University, Durham, NC; [8]Janssen Global Services LLC

BACKGROUND

- Acute limb ischemia (ALI) is a major adverse limb event (MALE) associated with high morbidity and mortality, and prior lower extremity revascularization (LER) is the greatest risk factor
- The VOYAGER PAD randomized trial examined PAD patients in the post LER setting. ALI was included in the primary endpoint.
- Data describing the presentation, initial care, and subsequent antithrombotic prophylaxis in patients with ALI after LER would provide valuable insights.

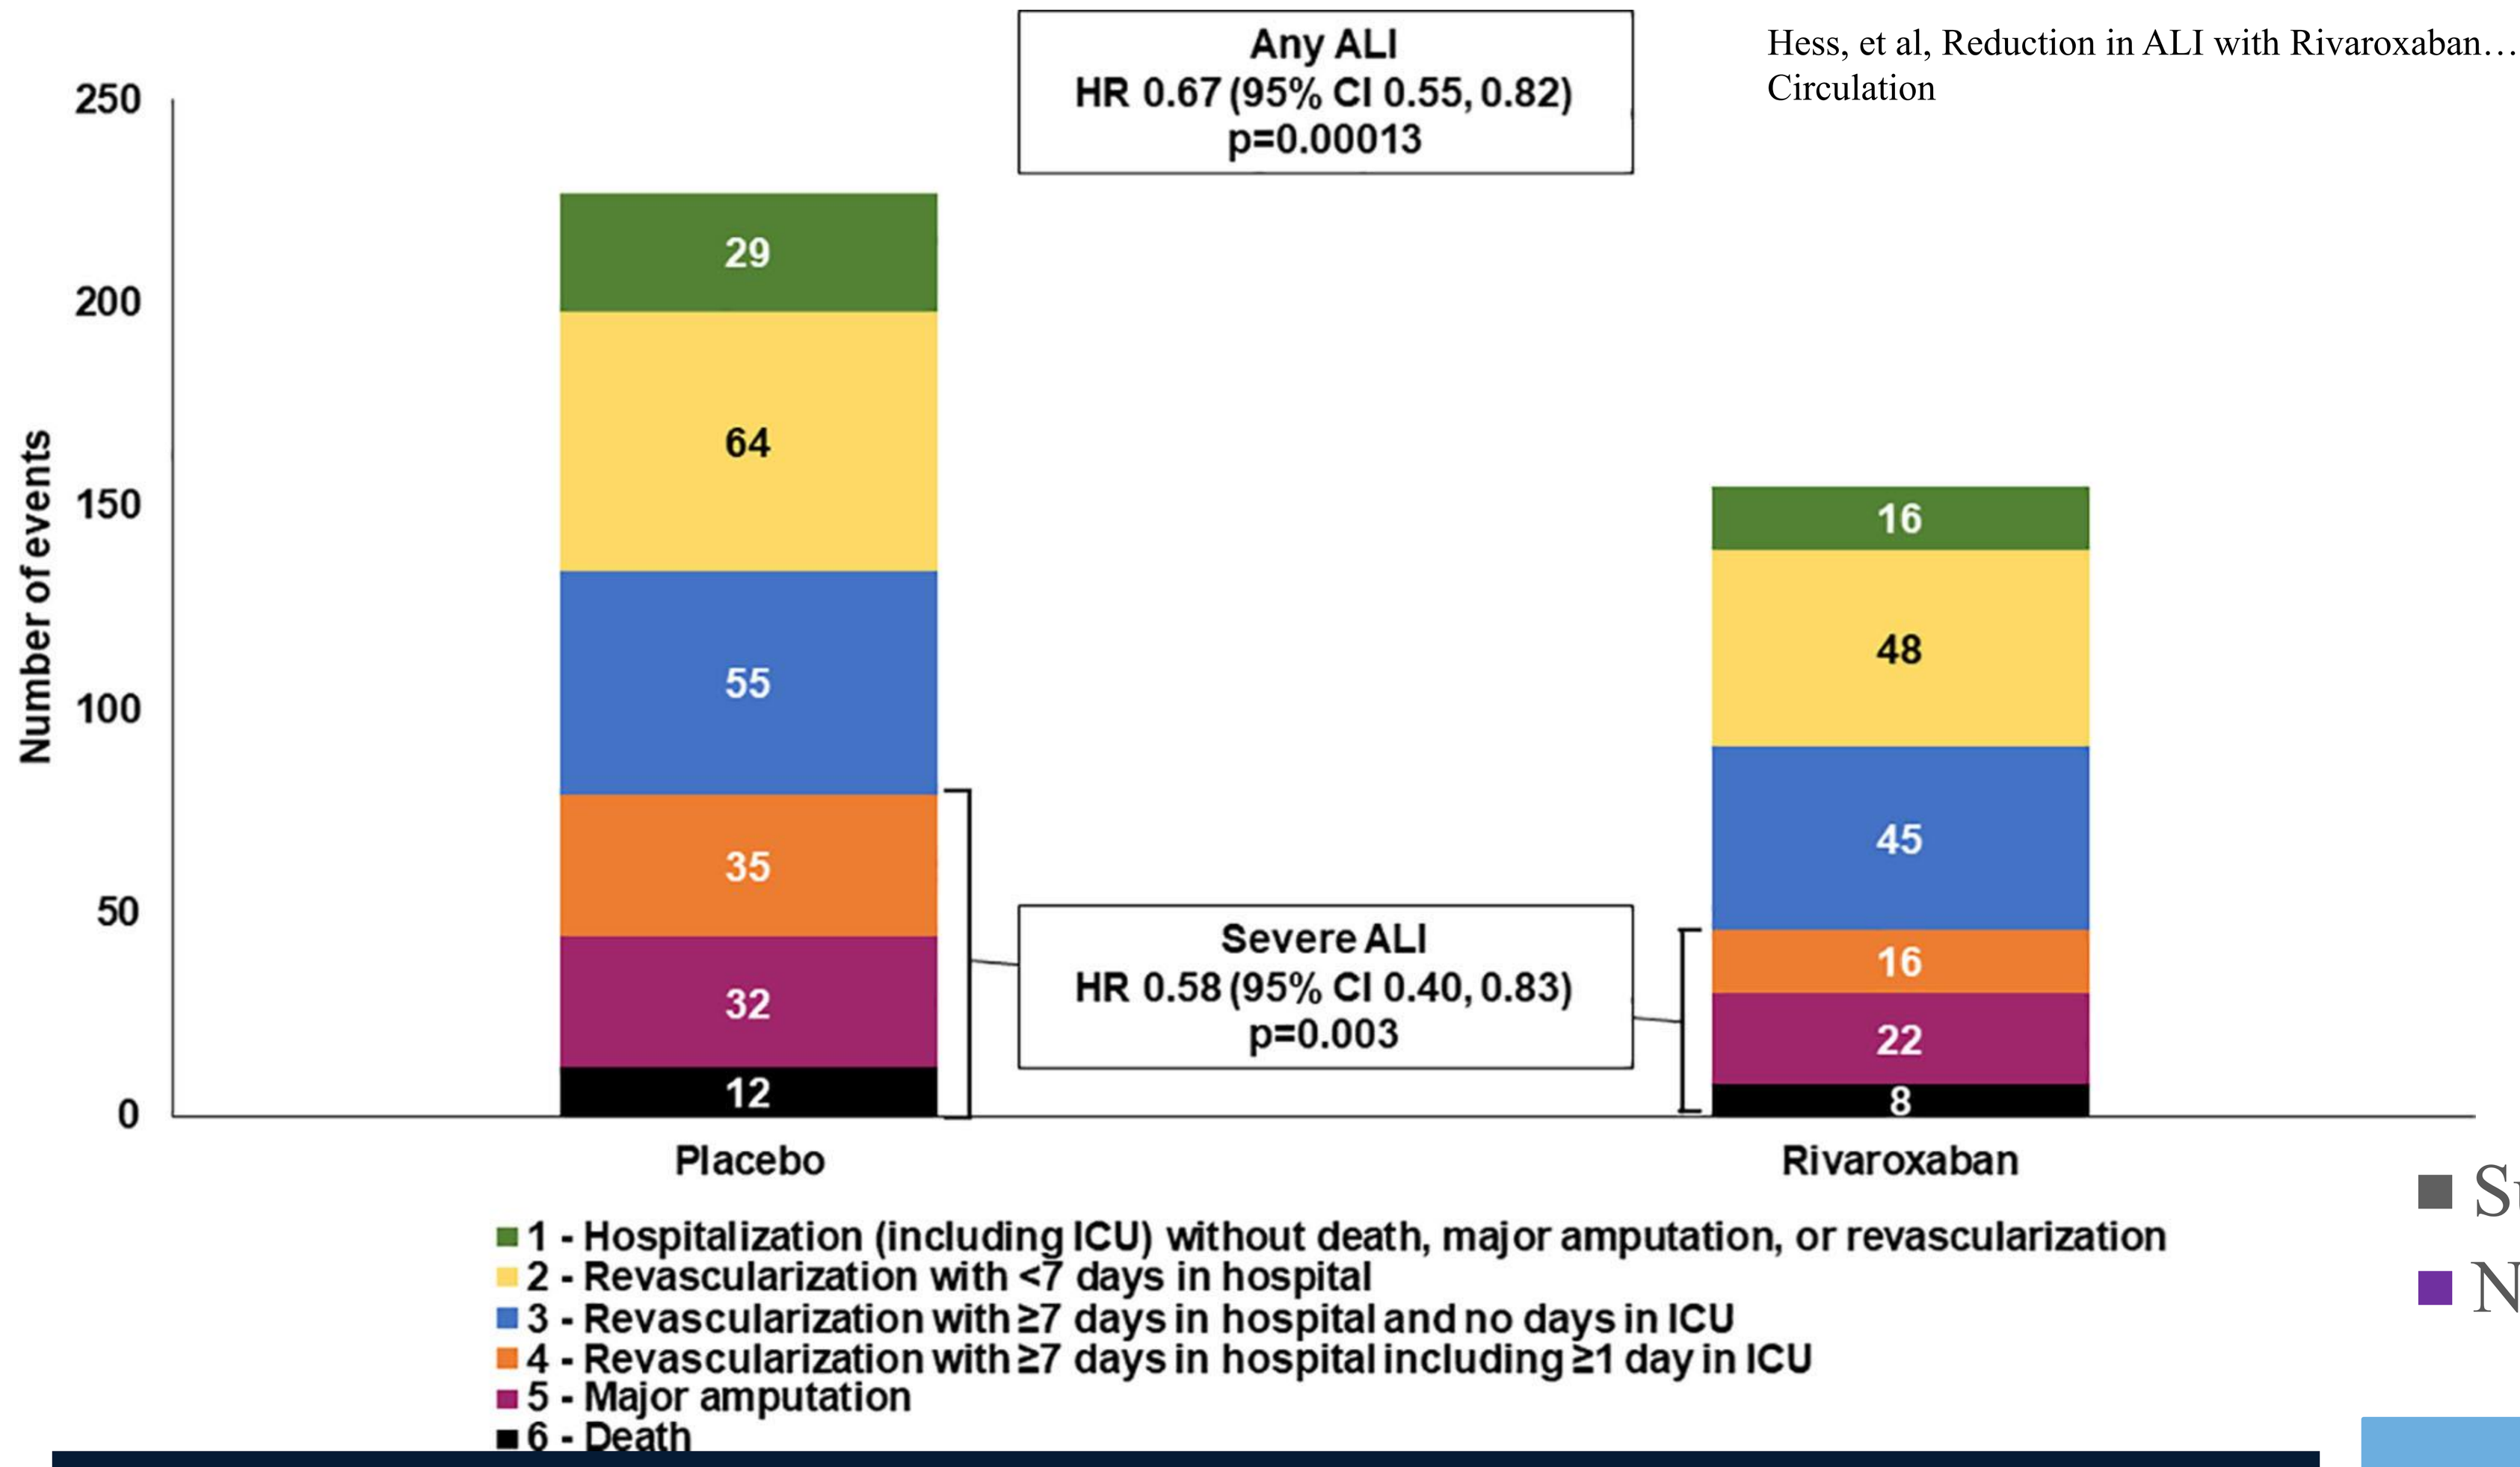

Any ALI
HR 0.67 (95% CI 0.55, 0.82)
p=0.00013
Severe ALI
HR 0.58 (95% CI 0.40, 0.83)
p=0.003
Number of events
250
200
150
100
50
0
29
64
55
35
32
12
16
48
45
16
22
8
Placebo
Rivaroxaban
1 - Hospitalization (including ICU) without death, major amputation, or revascularization
2 - Revascularization with <7 days in hospital
3 - Revascularization with ≥7 days in hospital and no days in ICU
4 - Revascularization with ≥7 days in hospital including ≥1 day in ICU
5 - Major amputation
6 - Death

Hess, et al, Reduction in ALI with Rivaroxaban… Circulation

METHODS

- The VOYAGER PAD trial randomized 6564 patients with PAD after LER to rivaroxaban 2.5 mg BID + ASA vs. placebo + ASA and followed for a median of 28 months
- Clinical dossiers for adjudicated ALI events reviewed by two trained vascular specialists who were blinded to treatment allocation for:
 – Etiology
 – Relationship to prior LER
 – Initial definitive intervention strategy
 – Hospitalization outcomes
 – Discharge antithrombotic pharmacotherapy

RESULTS

- 382 (5.8%) had an ALI event for a total of 508 events
- 155 (40.6%) of these patients who had ALI were randomized to rivaroxaban 2.5mg BID + ASA
- ~98% of all events were from in-situ thrombosis or artery to artery embolism and the vast majority at site of prior LER

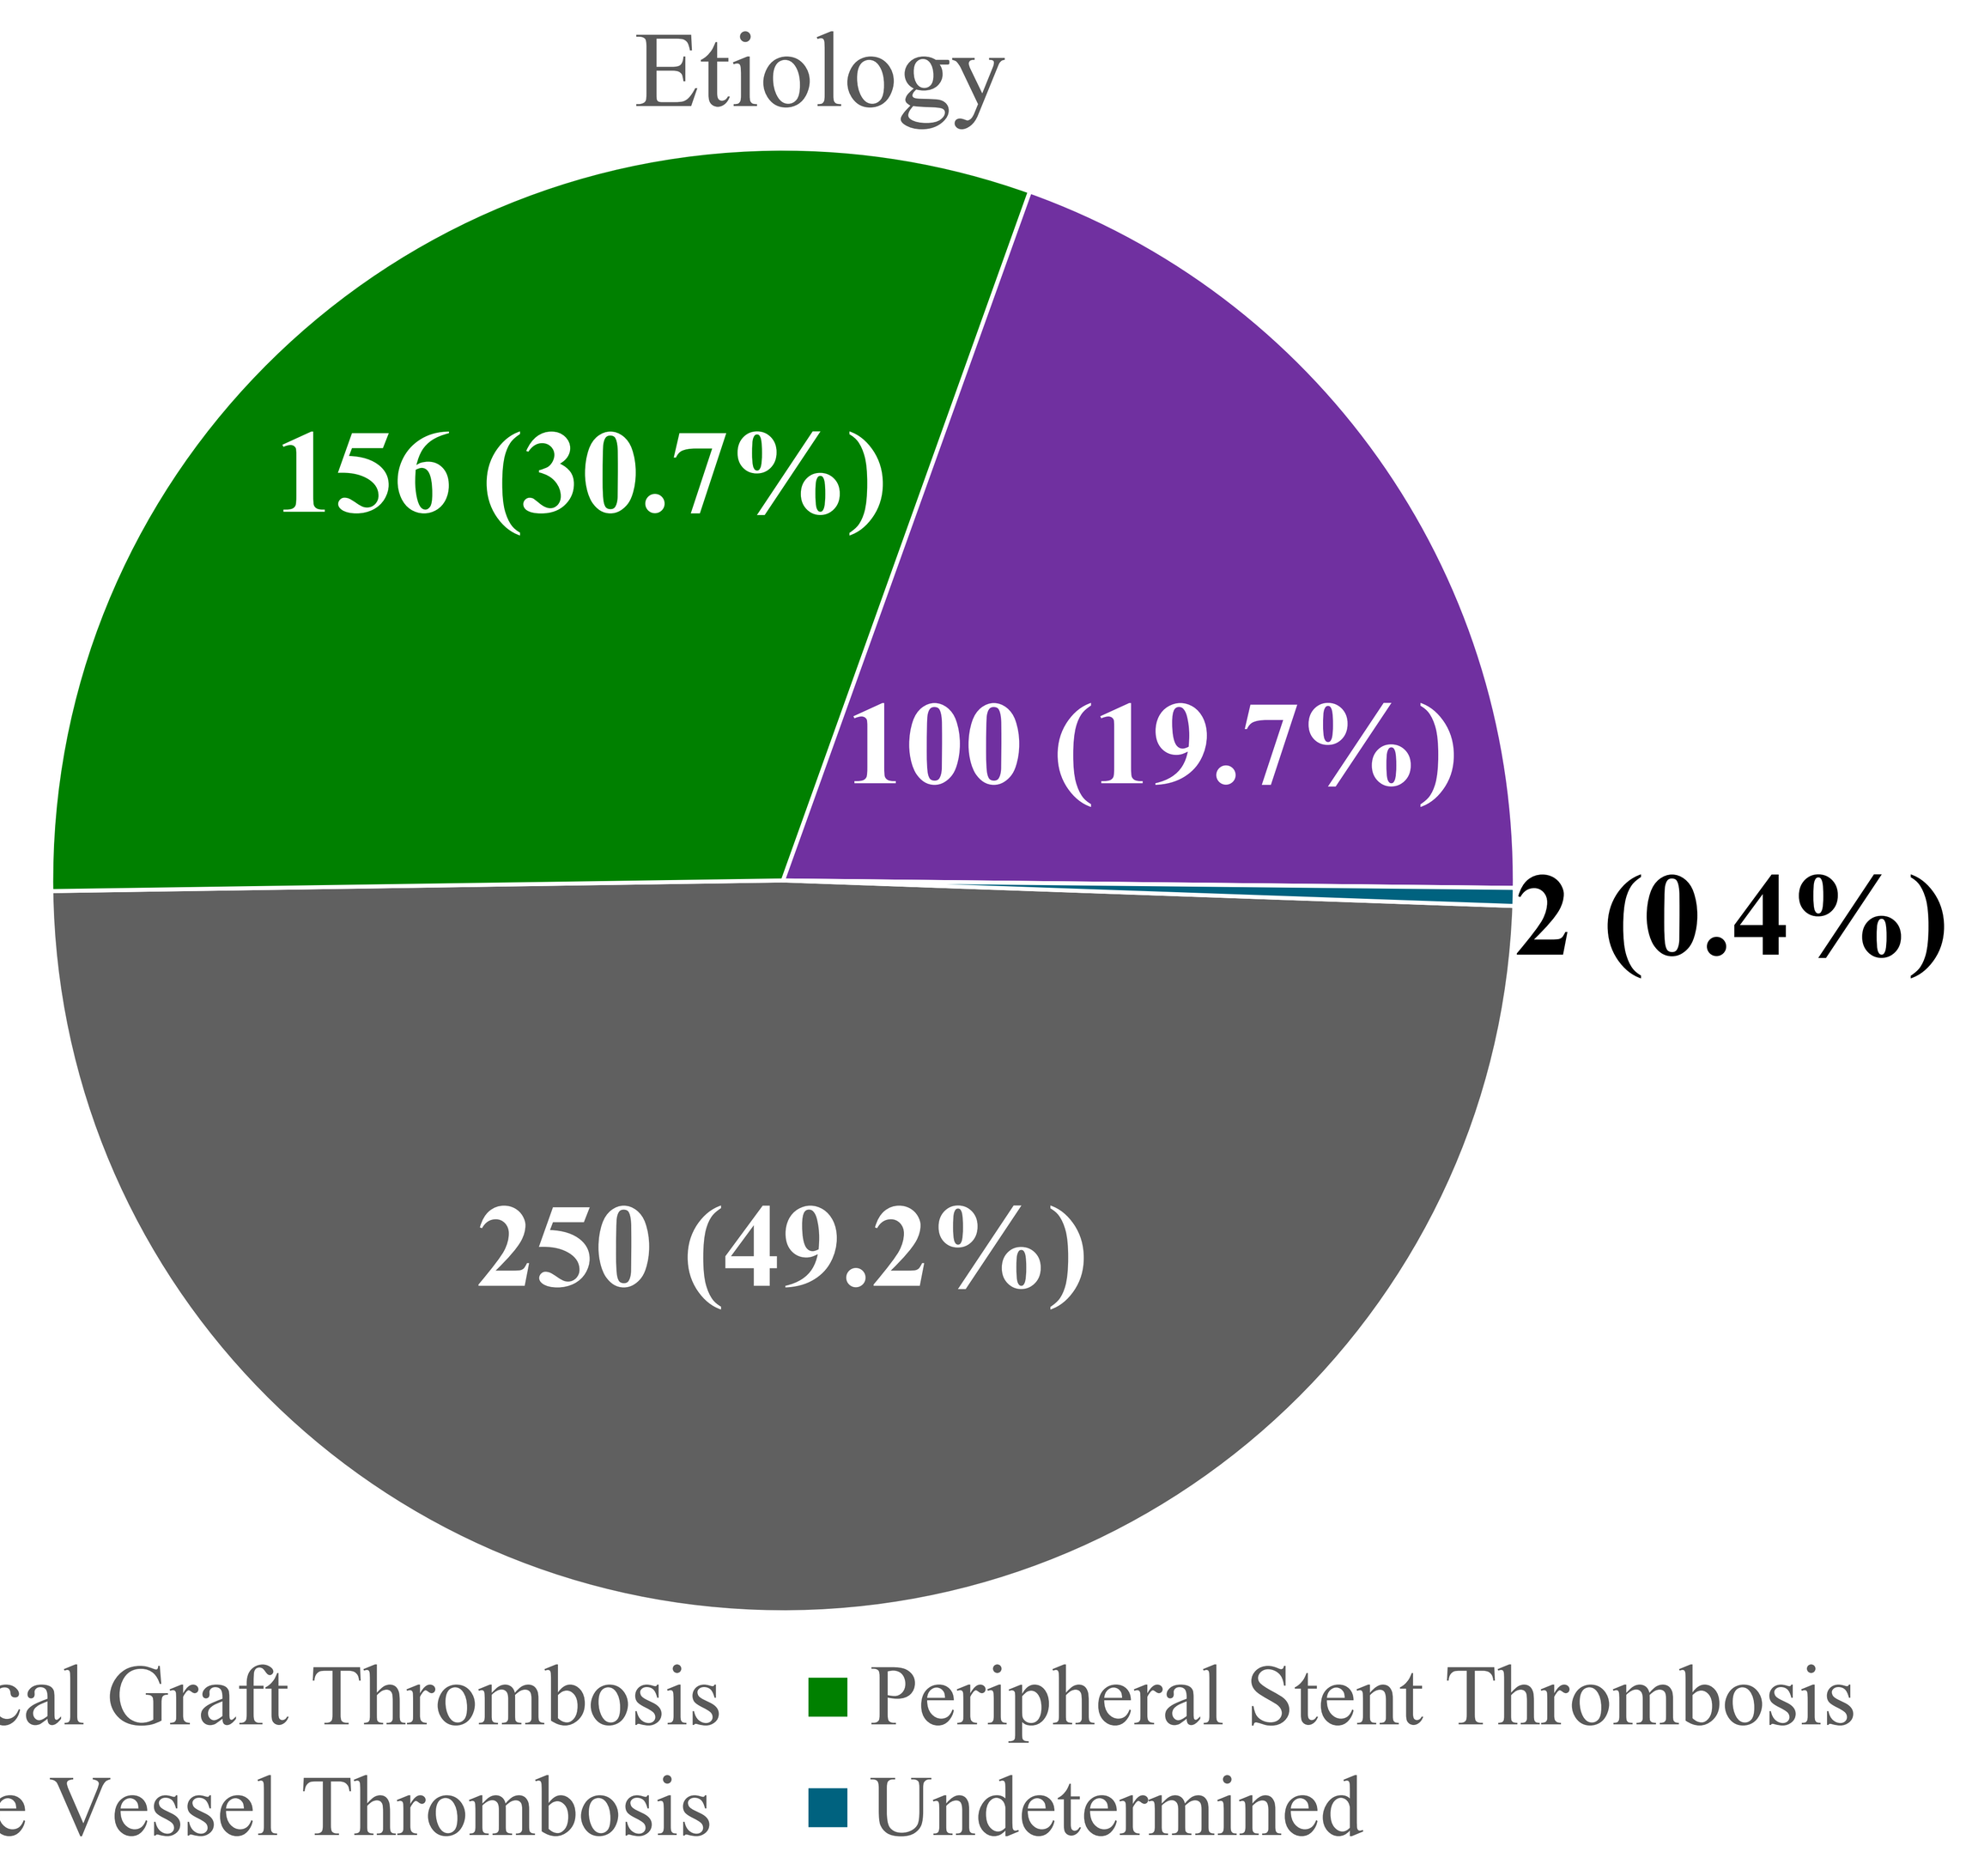

Etiology
156 (30.7%)
100 (19.7%)
2 (0.4%)
250 (49.2%)
Surgical Graft Thrombosis
Peripheral Stent Thrombosis
Native Vessel Thrombosis
Undetermined

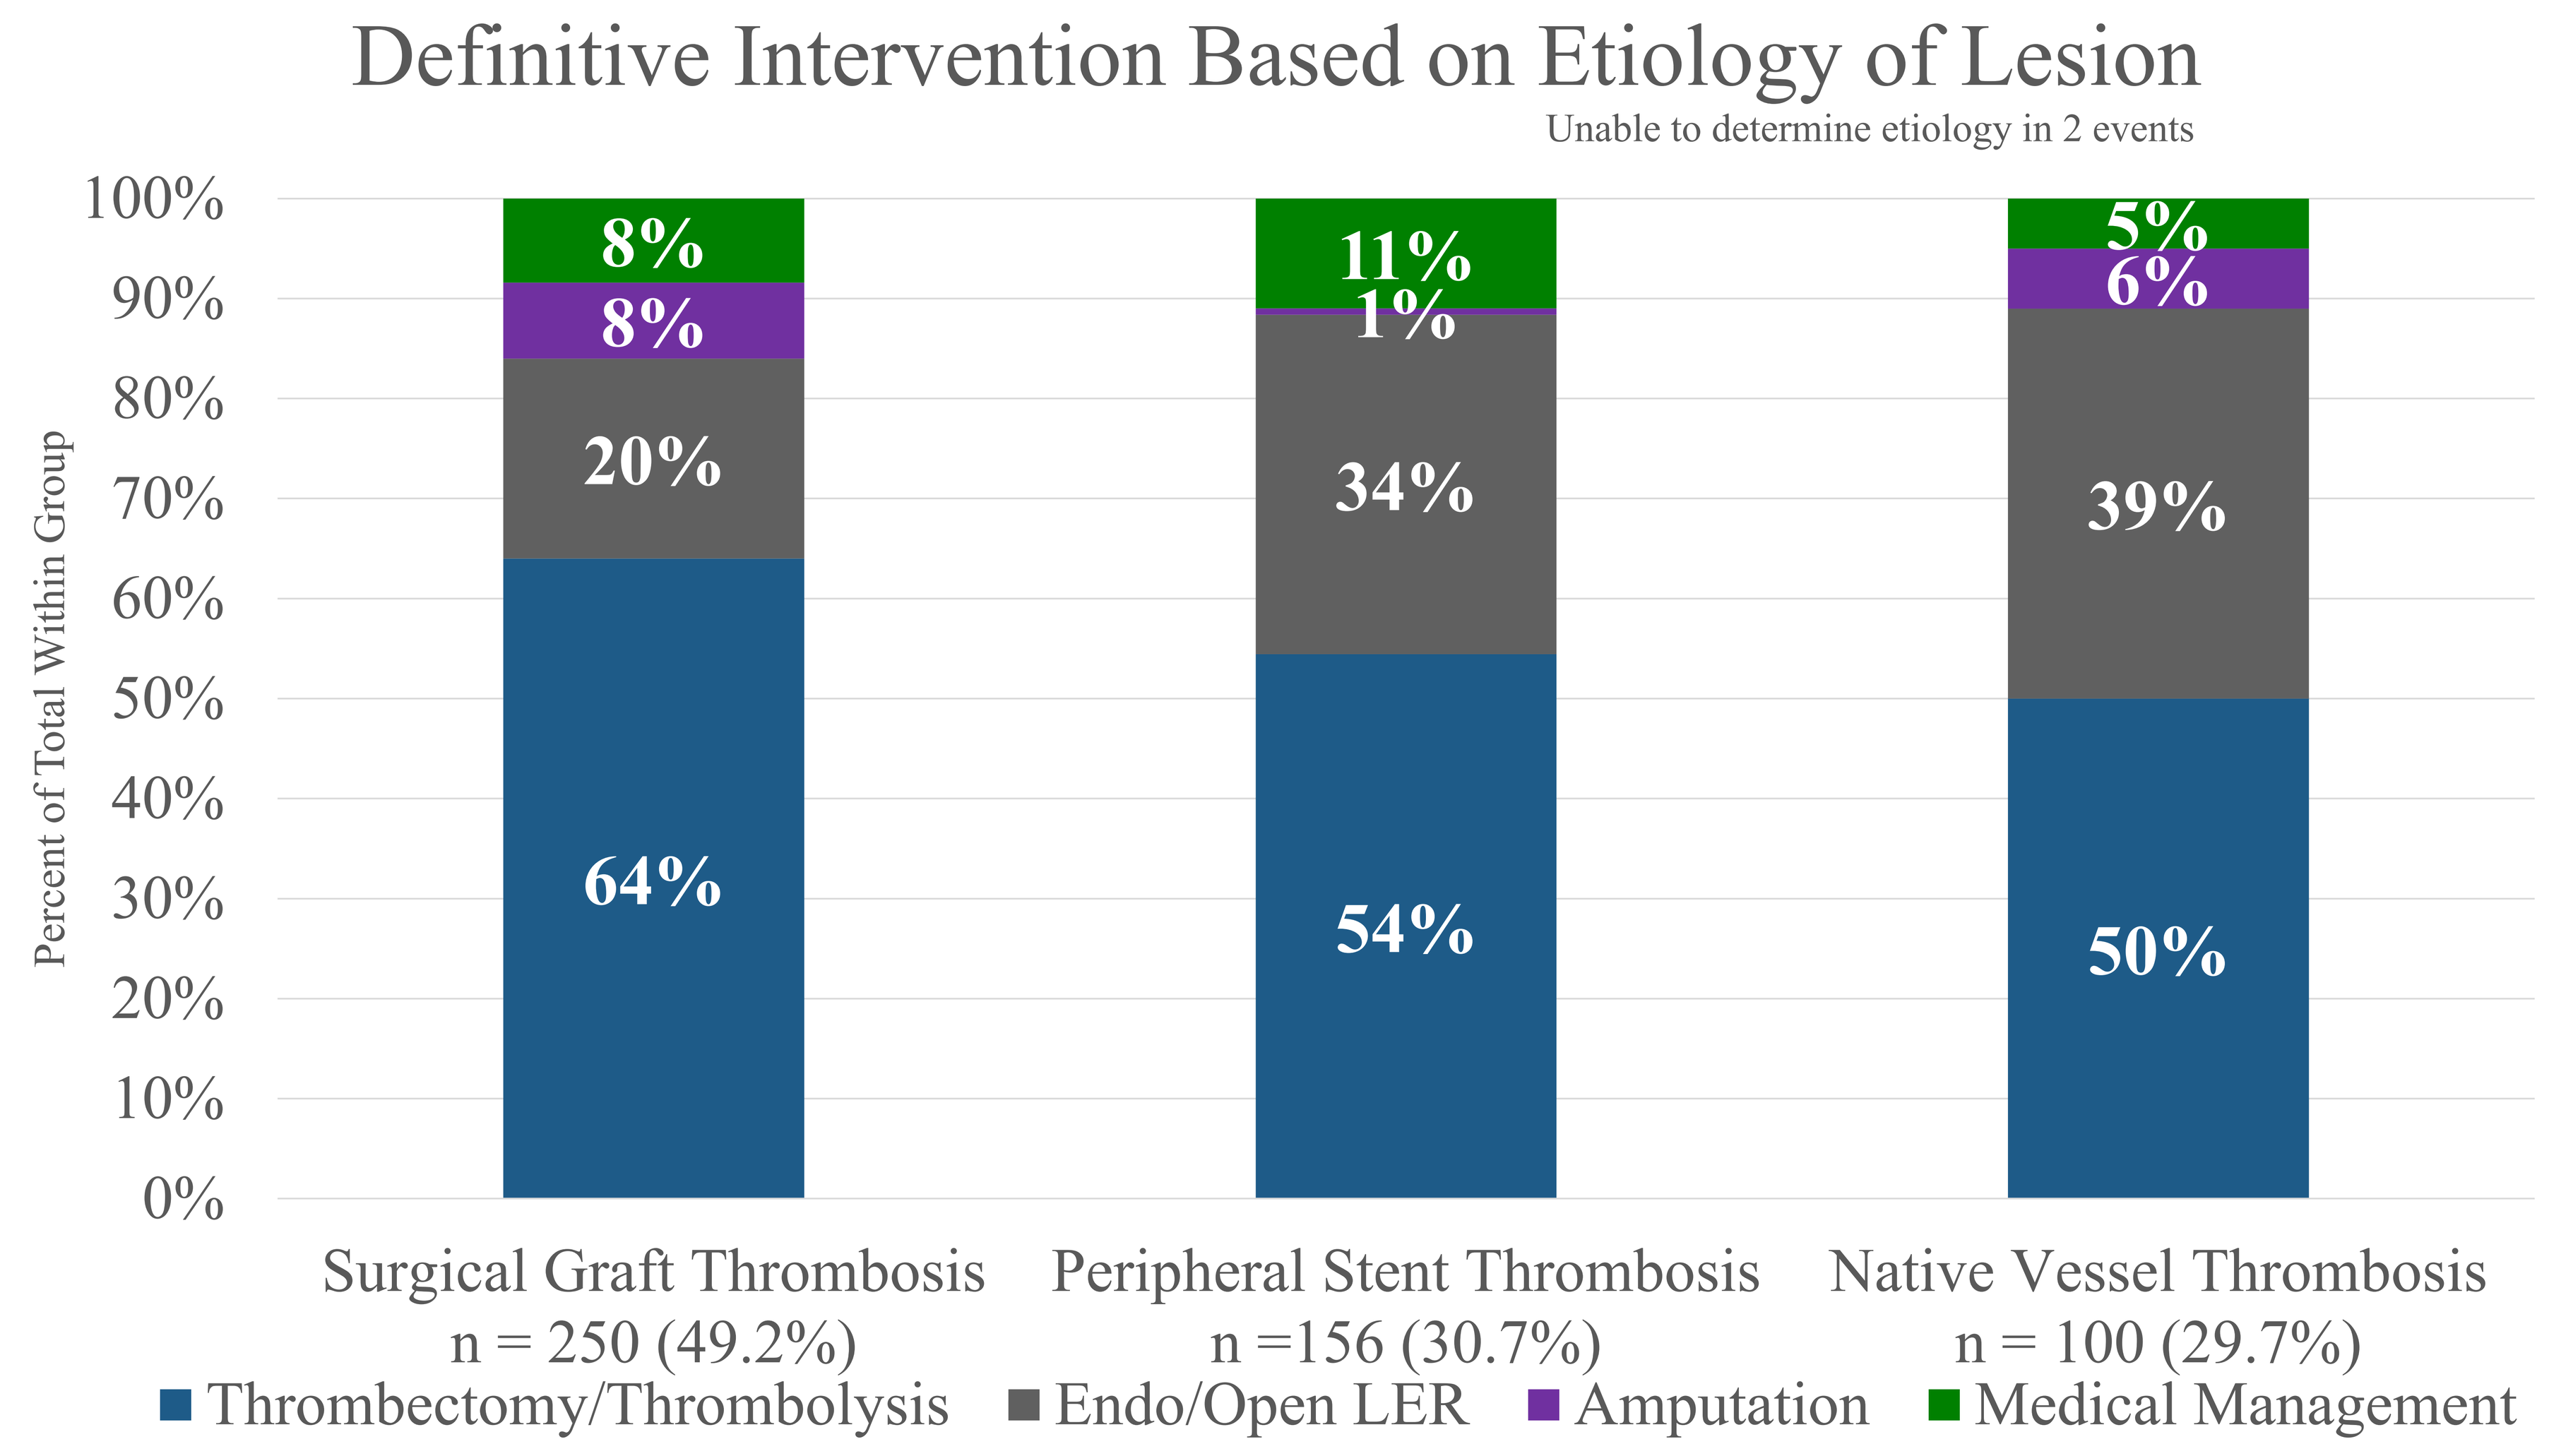

Definitive Intervention Based on Etiology of Lesion
Unable to determine etiology in 2 events
Percent of Total Within Group
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
8%
8%
20%
64%
11%
1%
34%
54%
5%
6%
39%
50%
Surgical Graft Thrombosis n = 250 (49.2%)
Peripheral Stent Thrombosis n =156 (30.7%)
Native Vessel Thrombosis n = 100 (29.7%)
Thrombectomy/Thrombolysis
Endo/Open LER
Amputation
Medical Management

Post Treatment Adverse Events

Number of Patients with Major AEs
- Fasciotomy: 29 (8%)
- Ventilator, shock, and/or dialysis 14 (4%)
- Major amputation: 54 (14.1%)
- Mortality: 20 (5.2%)

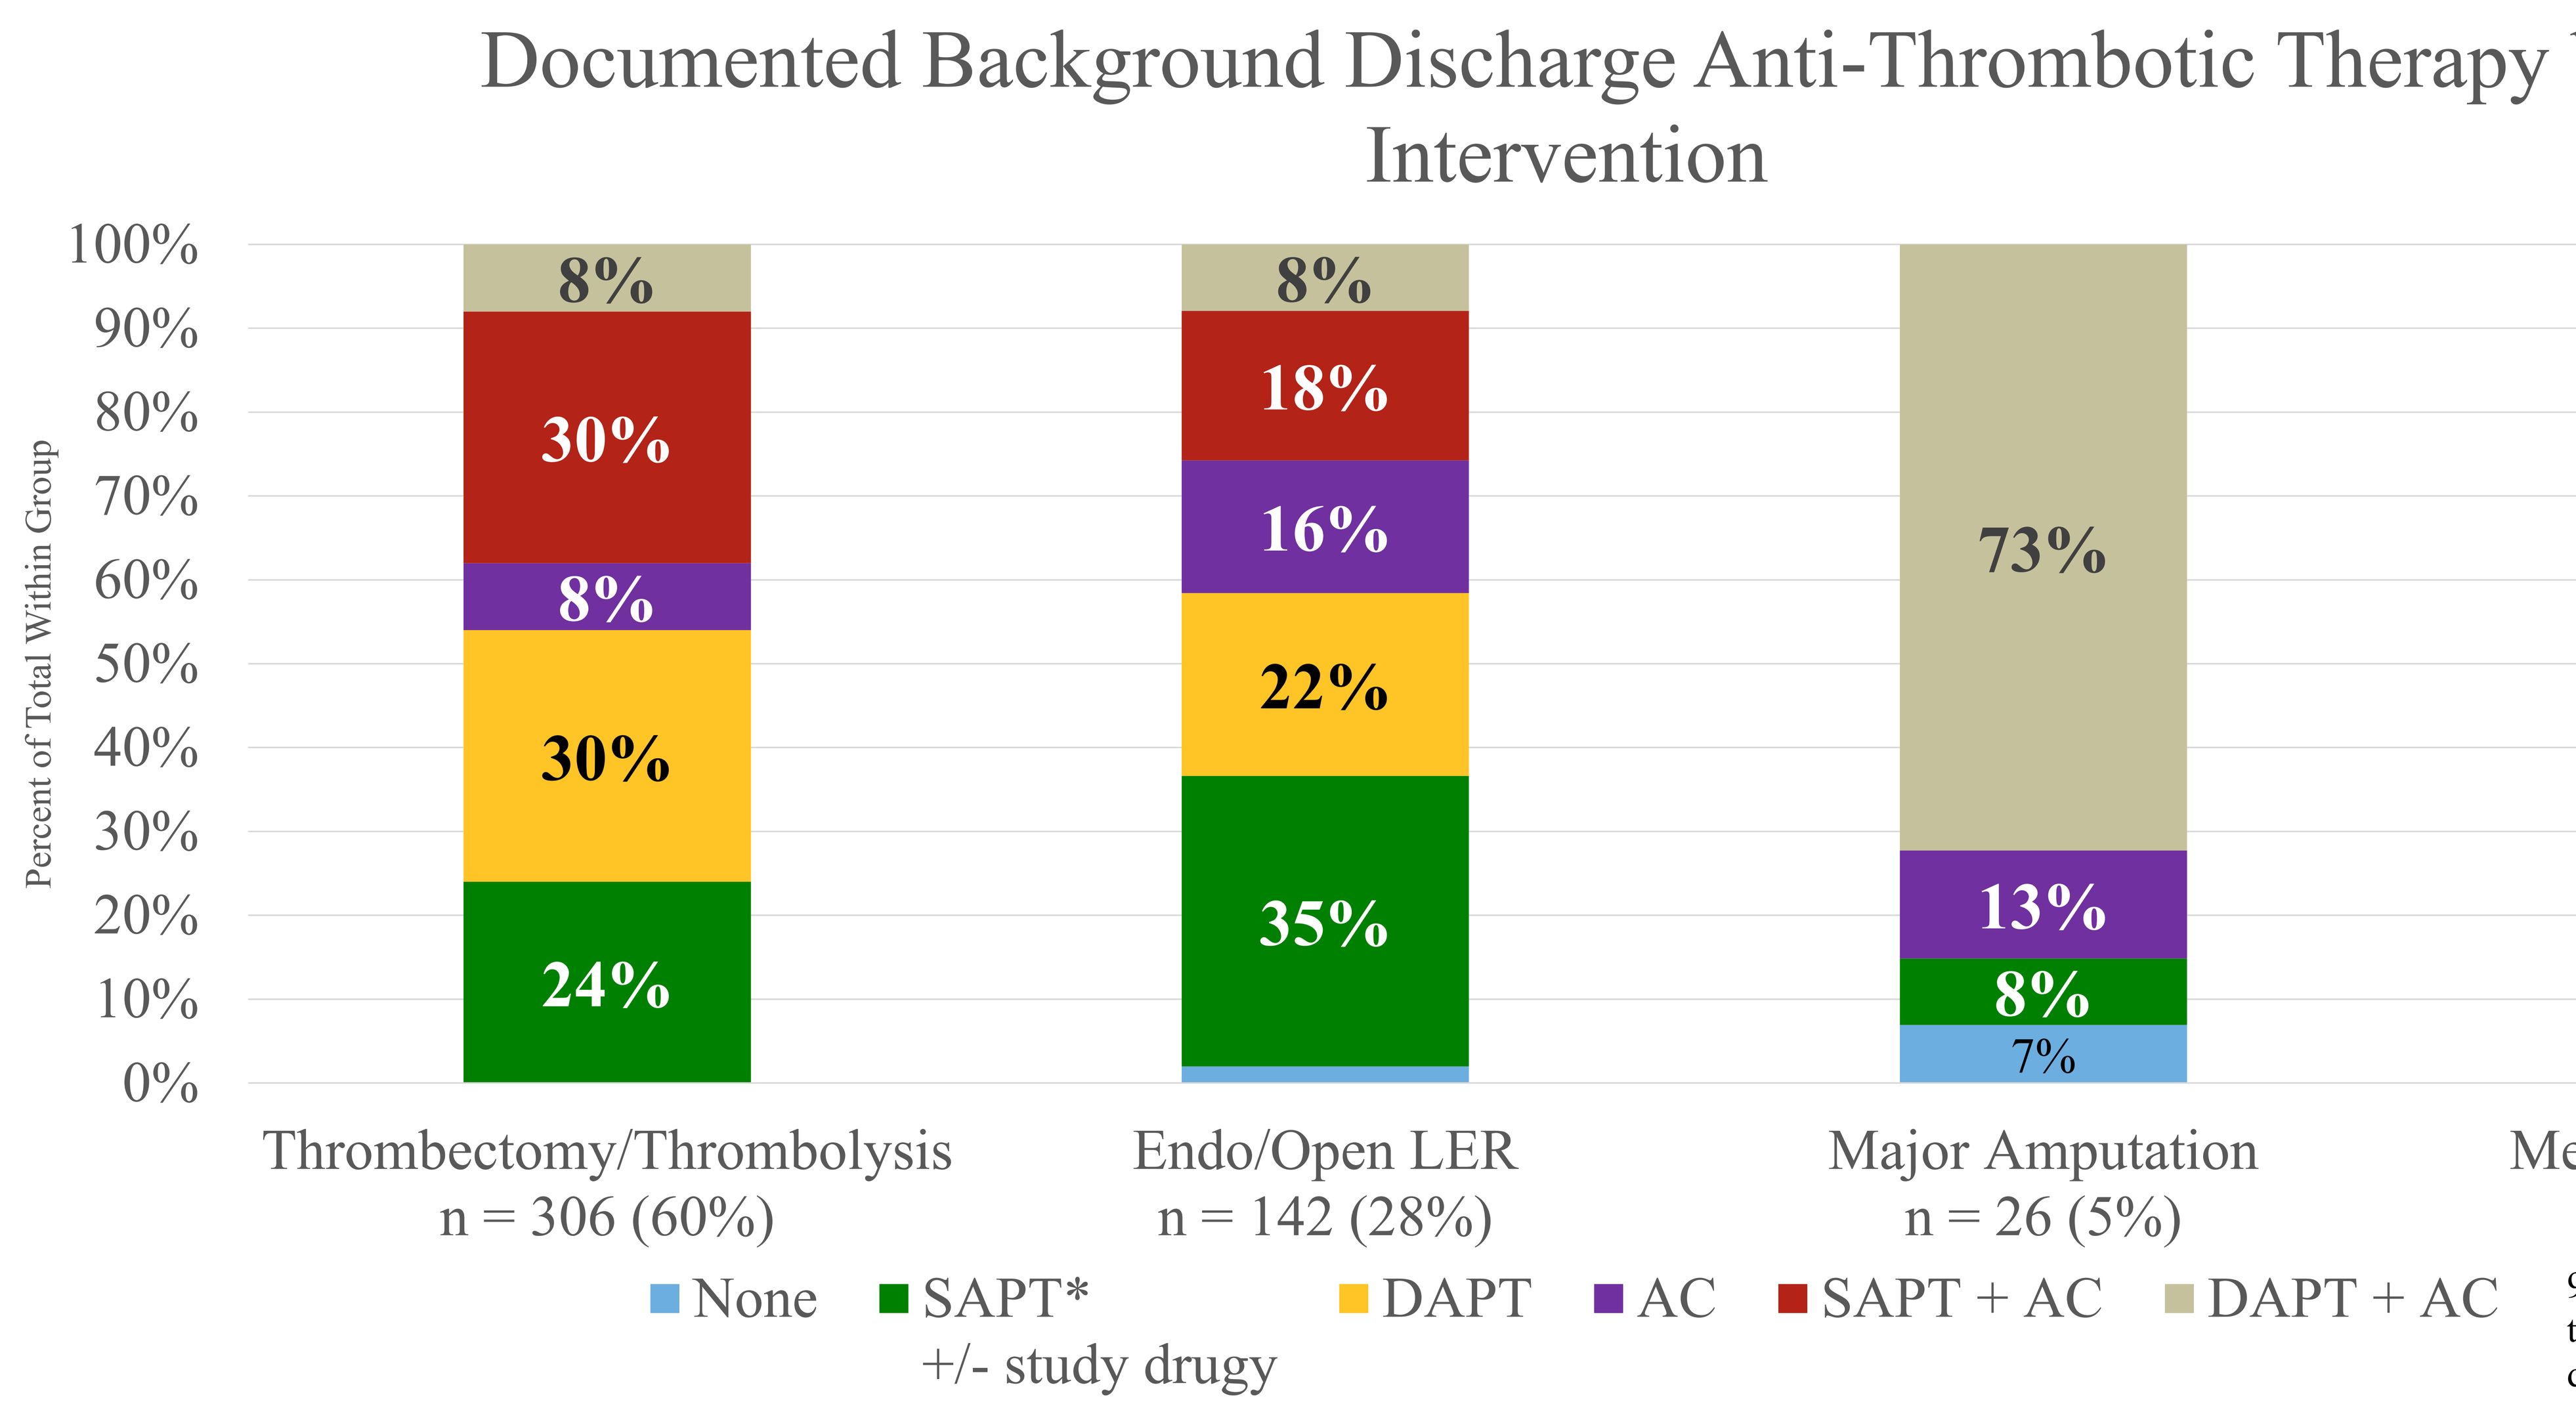

Documented Background Discharge Anti-Thrombotic Therapy by Intervention
Percent of Total Within Group
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
8%
30%
8%
30%
24%
8%
18%
16%
22%
35%
73%
13%
8%
7%
8%
4%
12%
72%
4%
Thrombectomy/Thrombolysis n = 306 (60%)
Endo/Open LER n = 142 (28%)
Major Amputation n = 26 (5%)
Medical Management n = 34 (17%)
None
SAPT* +/- study drugy
DAPT
AC
SAPT + AC
DAPT + AC

97/508 (19%) had no discharge anti-thrombotics documented in the event dossiers

CONCLUSIONS

- ALI in the post-LER setting is frequent
- The etiology of the vast majority of ALI events are either surgical graft or peripheral stent thrombosis
- The most common procedure performed for ALI is a thrombectomy/thrombolysis followed by an endo/open LER regardless of the etiology of the event
- Discharge anti-thrombotic strategy is varied regardless of type of intervention

IMPLICATIONS

- Few randomized studies have evaluated optimal acute treatment and post-event prevention for patients presenting with ALI
- In a modern, well characterized cohort of patients with PAD, including 508 ALI events, treatments were highly variable
- Future studies evaluating optimal treatment approaches to ALI may lead to an evidence base informing more systematic approaches to this severe event

DISCLOSURES

VOYAGER was designed and overseen by a collaborative group that included Colorado Prevention Center (CPC) Clinical Research (an academic research organization affiliated with the University of Colorado), the academic executive committee and the sponsors, Bayer and Janssen Pharmaceuticals. JH reports owning AstraZeneca stock and research funding to CPC Clinical Research from Arca Biopharma and Janssen. SA reports lecture fees from Bayer and Janssen. MRN, WHC, LD - no disclosure. MRP reports grant support, advisory board fees and consulting fees from AstraZeneca, grant support from Medtronic and Philips Healthcare, and grant support and advisory board fees from Heartflow. WRH reports grant support from Amgen and AstraZeneca. SD reports grant support from Cook and Terumo Aortic. CNH reports research funding to CPC Clinical Research from Merck, Bayer, and Amgen. SDB and EM are employed by Bayer. LPH is employed by Janssen Pharmaceuticals and owns stock in Johnson & Johnson RMB reports consulting fees and lecture fees from Bristol-Myers Squibb, Daiichi Sankyo. MPB reports research grants to CPC Clinical Research from Alnylam, Amgen, Arca, AstraZeneca, Bayer, CellResearch, Eidos, NovoNordisk, Osiris, Terumo