Newer guideline-directed medical therapies are underutilized in 2021-2 in patients with heart failure and chronic kidney disease

An Affiliate of:

Mario Enrico Canonico MD[1,2], Judith Hsia MD[1,2], Christopher P. Cannon MD[3], Marc P. Bonaca MD, MPH[1,2]

[1]CPC Clinical Research, Aurora, CO, USA, [2]Department of Medicine, University of Colorado, Aurora, CO, USA, [3]Cardiovascular Division, Brigham & Women's Hospital, Boston, MA, USA

An Affiliate of:

BACKGROUND

Chronic kidney disease (CKD) is a common comorbidity in patients with heart failure (HF). In 2021 and 2022, HF guidelines endorsed newer therapies such as angiotensin receptor-neprilysin inhibitors (ARNI) and sodium-glucose cotransporter-2 inhibitors (SGLT2i) as first-line medications to improve clinical outcomes in HF patients with or without CKD (Table 1) (1,2). Our analysis evaluated the hypothesis that prescription of guideline directed medical therapy (GDMT) is similar in HF patients with or without CKD.

Table 1. ACC/AHA and ESC guidelines on Heart Failure medications.

	HFrEF	HFpEF
ESC 2021	• ACEi/ARB/ARNI (Class I) • Beta Blocker (Class I) • MRA (Class I) • SGLT2i (Class I) • Loop Diuretic (Class I) • CVD management (Class I)	• CVD management (Class I) • Diuretics as needed (Class I) • SGLT2i with DM and high CV risk (Class I)
ACC/AHA 2022	• ACEi/ARB/ARNI (Class I) • Beta Blocker (Class I) • MRA (Class I) • SGLT2i (Class I) • Loop Diuretic (Class I) • CVD management (Class I)	• CVD management (Class I) • Diuretics as needed (Class I) • SGLT2i (Class IIa) • ARB/ARNI (Class IIb) • MRA (Class IIb)

HFrEF= Heart Failure with Reduced Ejection Fraction; HFpEF= Heart Failure with Preserved Ejection Fraction; ESC= European Society of Cardiology; ACC/AHA= American College of Cardiology/American Heart Association; DM= Diabetes mellitus; CVD= Cardiovascular disease.

METHODS

We extracted de-identified data from TriNetX for adults with healthcare encounters with a diagnosis of HF from May 2021-April 2022 in the University of Colorado health system using HF ICD-10 codes.

CKD stages were defined by estimated glomerular filtration rate (eGFR) according to (Kidney Disease Improving Global Outcomes) guidelines. The Colorado Multiple Institutional Review Board provided a waiver of informed consent. Categories were compared by chi-square.

RESULTS

Among 17990 patients with HF encounters, 51% had reduced (HFrEF) and 49% preserved ejection fraction (HFpEF). Mild CKD was more common among patients with HFrEF than HFpEF (Stage G1 25% vs 20%, $p<0.0001$; G2 40% vs 38%, $p<0.05$) while more severe CKD was more common in patients with HFpEF (G3 32% vs 37%, $p<0.0001$; G4 4% vs 5%, $p=0.0005$). Older GDMT (beta-blockers, ACEi/ARB) were widely prescribed irrespective of HF phenotype and CKD stage (Figure 1,2). Use of mineralocorticoid receptor antagonists was intermediate (despite Finerenone resulted not prescribed in our cohort), whereas use of newer GDMT (ARNI and SGLT2i) was less frequent in both HFrEF and HFpEF (Figure 1).

Figure 1. GDMT in HFrEF and HFpEF patients based on CKD stage.

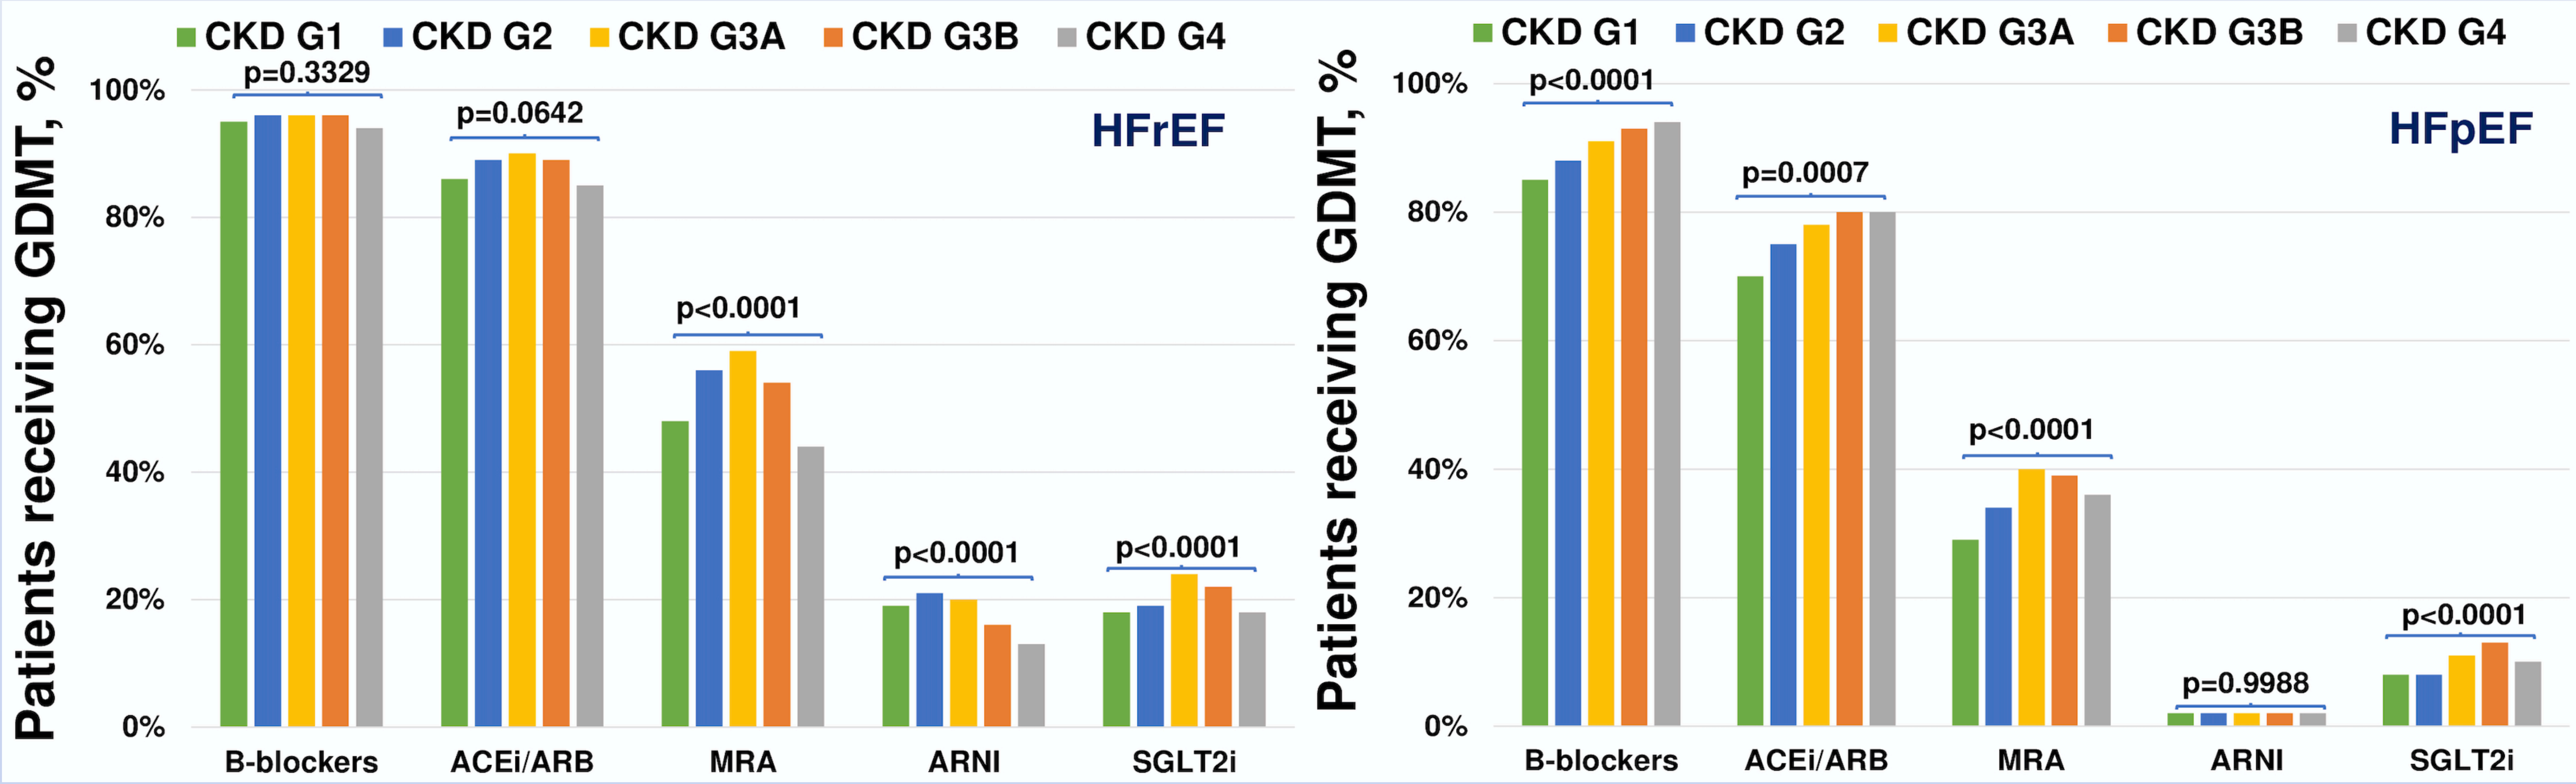
CKD G1
CKD G2
CKD G3A
CKD G3B
CKD G4
HFrEF
Patients receiving GDMT, %
100%
80%
60%
40%
20%
0%
p=0.3329
p=0.0642
p<0.0001
p<0.0001
p<0.0001
B-blockers
ACEi/ARB
MRA
ARNI
SGLT2i
HFpEF
p<0.0001
p=0.0007
p<0.0001
p=0.9988
p<0.0001

DISCUSSION

HF patients with CKD are at increased risk for cardiovascular death and hospitalization for HF (3). Despite HF updated guidelines, use of GDMT remained suboptimal even in 2021-22 in a large and diverse health system. While older medications as beta-blockers, ACEi/ARB and MRA resulted widely prescribed, the uptake of newer agents appears not yet optimal. Clinical inertia, knowledge gaps, cost, practitioner and patient fear of adverse effects could represent concurrent causes.

LIMITATIONS

The analysis was conducted on retrospective and pooled observational data which lacked patient level information.

CONCLUSION

- ARNI and SGLT2i use even in 2021-2 was infrequent among patients with HF irrespective of CKD stage. Use of either type MRA is intermediate in this population of HF and CKD.
- Efforts to accelerate uptake of newer guideline-directed medical therapies are needed to improve HF outcomes, irrespective of CKD stage and HF phenotype.

REFERENCES

1) McDonagh TA, et al ESC Scientific Document Group. 2021 ESC Guidelines for the diagnosis and treatment of acute and chronic heart failure. Eur Heart J. 2021 Sep 21;42(36):3599-3726.
2) Heidenreich PA, et al. 2022 AHA/ACC/HFSA Guideline for the Management of Heart Failure: A Report of the American College of Cardiology/American Heart Association Joint Committee on Clinical Practice Guidelines. Circulation. 2022 May 3;145(18):e895-e1032.
3) Bhatt DL, et al. Sotagliflozin in Patients with Diabetes and Chronic Kidney Disease. N Engl J Med. 2021 Jan 14;384(2):129-139.

FINANCIAL DISCLOSURES

This project was supported by a grant from Lexicon Pharmaceuticals, The Woodlands, TX, USA.
JH, MPB receive salary support through their universities from CPC Clinical Research, a non-profit academic research organization affiliated with the University of Colorado. CPC receives research grants from Amgen, Better Therapeutics, Boehringer-Ingelheim (BI), Bristol-Myers Squibb (BMS), Daiichi Sankyo, Janssen, Merck, Novo Nordisk, Pfizer. Consulting fees from Aegerion/Amryt, Alnylam, Amarin, Amgen, Applied Therapeutics, Ascendia, BI, BMS, Eli Lilly, Janssen, Lexicon, Merck, Pfizer, Rhoshan, Sanofi. He serves on Data and Safety Monitoring Boards for the Veteran's Administration, Applied Therapeutics and NovoNordisk.

APPRISOR™