HFSA 2023 ANNUAL SCIENTIFIC MEETING
WHERE HEART FAILURE TEAMS GATHER

How generalizable is the SOLOIST-WHF trial in ‘real world’ patients with diabetes admitted with heart failure? Results from a large US integrated health system

Mario Enrico Canonico[1,2], **Judith Hsia**[1,2], **Michael Szarek**[1,2,3], **Christopher P. Cannon**[4], **Marc P. Bonaca**[1,2]

[1] CPC Clinical Research, Aurora, CO, USA; [2] Department of Medicine, University of Colorado, Aurora, CO, USA; [3] Downstate School of Public Health, State University of New York, NY, USA; [4] Cardiovascular Division, Brigham & Women's Hospital, Boston, MA, USA

CPC Clinical Research — An Affiliate of: CU

INTRODUCTION

In the Sotagliflozin in Patients with Diabetes and Recent Worsening Heart Failure (SOLOIST-WHF) trial the sodium-glucose cotransporter 1/2 inhibitor (SGLT1/2) sotagliflozin reduced the total number (first and subsequent) of hospitalizations or urgent visits for heart failure (HF) and cardiovascular death by 33% among patients with type 2 diabetes mellitus (T2DM) and recent hospitalization for heart failure (HHF) (Figure 1).[1]

Figure 1. Rates of the primary efficacy endpoint in SOLOIST-WHF.

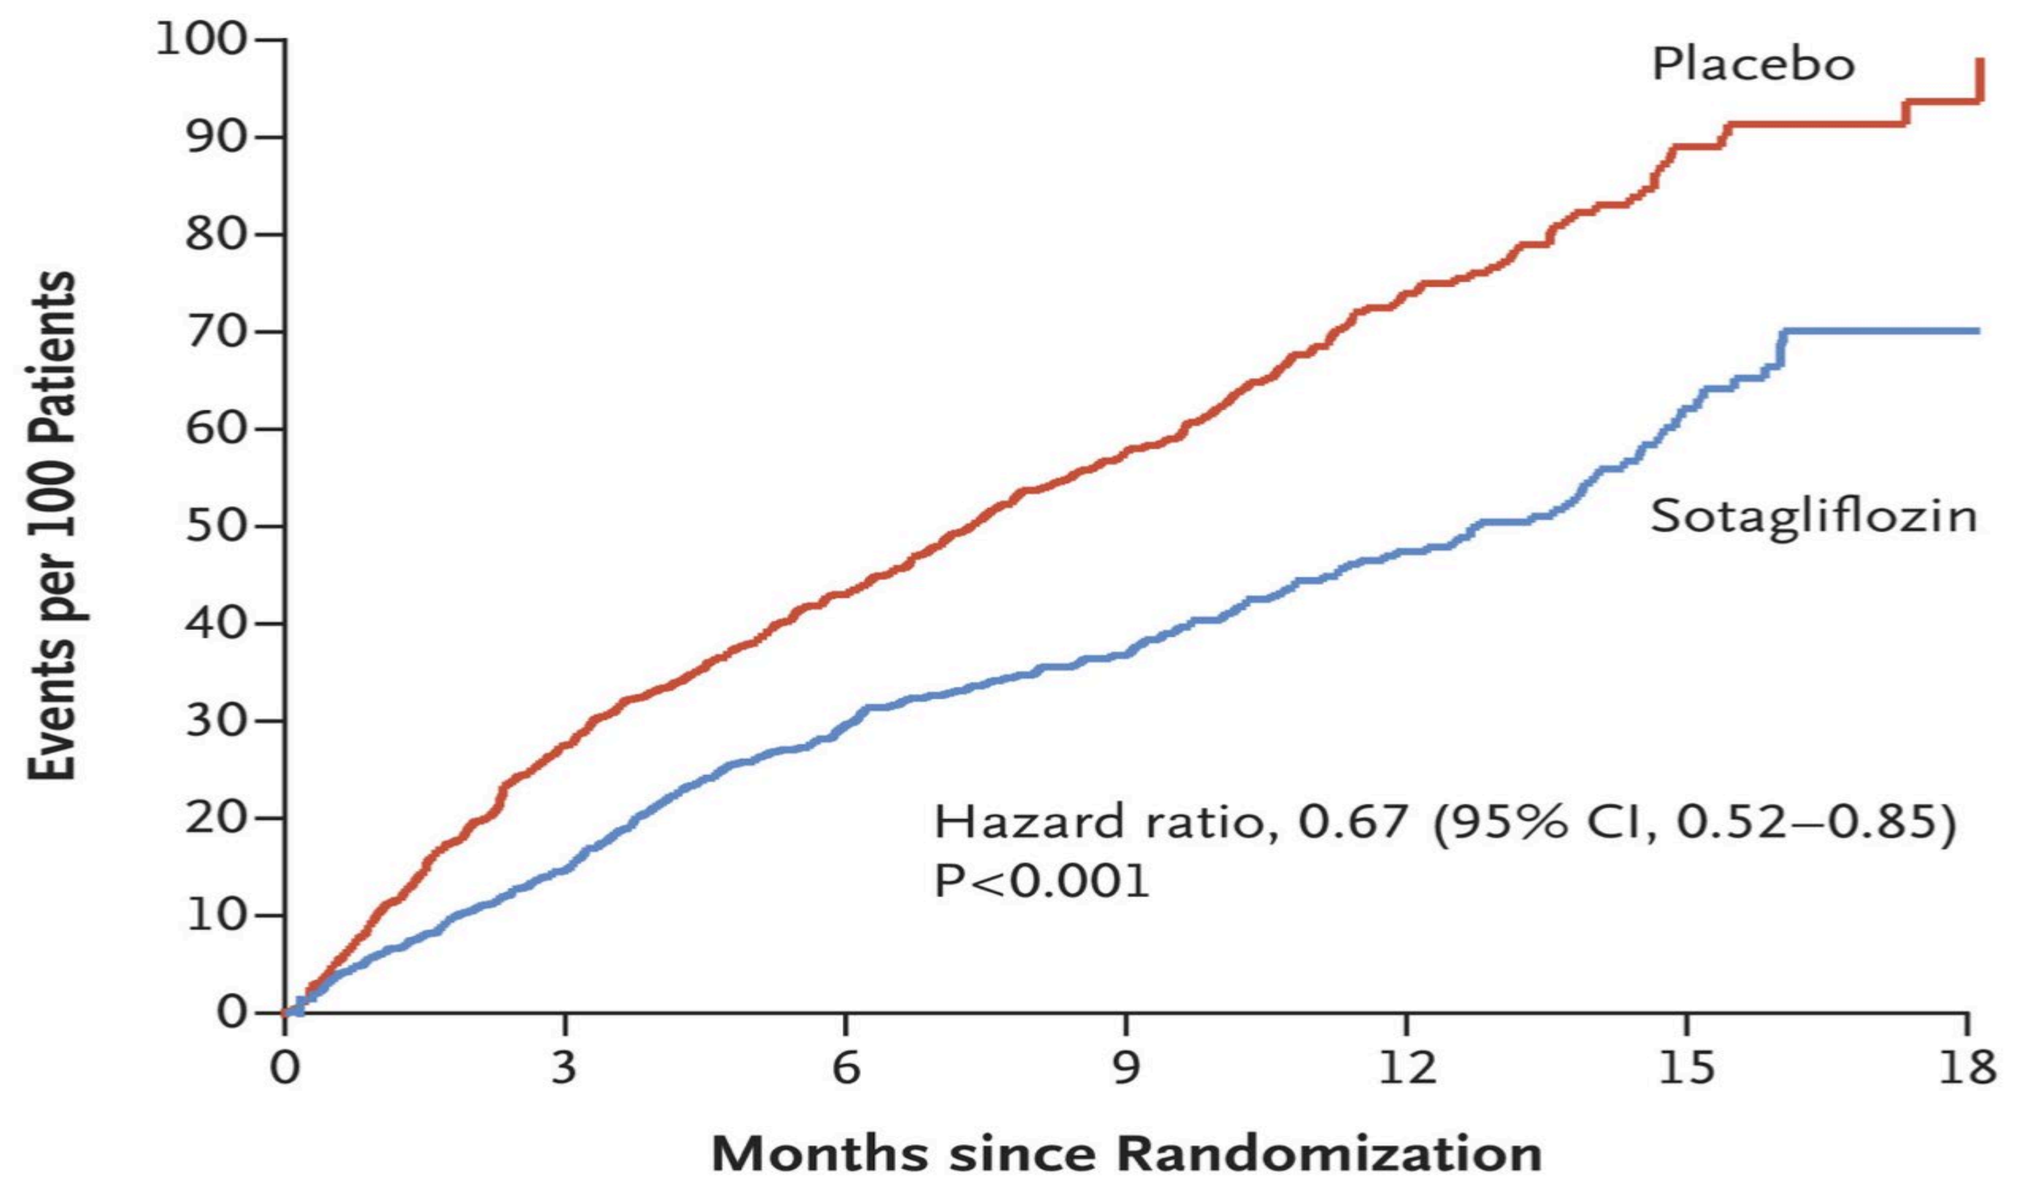
Placebo
Sotagliflozin
Hazard ratio, 0.67 (95% CI, 0.52–0.85)
P<0.001
Events per 100 Patients
Months since Randomization
0
3
6
9
12
15
18
0
10
20
30
40
50
60
70
80
90
100

AIM AND METHODS

- The aim of the abstract was to assess the generalizability of the SOLOIST-WHF trial in a real-world setting of patients affected by T2DM and admitted with HF in 2022.
- A SOLOIST-like group of patients was compared versus a group not meeting trial criteria from a large, diverse and integrated health system including urban and rural patients (Table 1).
- Patients in the University of Colorado health system TriNetX database were selected by International Classification of Diseases (ICD) codes as previously described.[2]
- Baseline characteristics of two groups were compared by chi-squared or t-test. One-year survival was compared by Kaplan-Meier and proportional hazards analyses.

Table 1. Selection criteria of the SOLOIST-like group.

Inclusion criteria	Exclusion criteria
HFrEF or HFpEF	Age <18 years or >85 years
Type 2 DM	Acute coronary syndrome < 3 months
Inpatient encounter or emergency visit	Stroke < 3 months
Loop diuretic	Dialysis < 1 year
	End stage heart failure
	Chronic kidney disease stage IV-V

HFrEF: heart failure with reduced ejection fraction; HFpEF: heart failure with preserved ejection fraction; DM: diabetes mellitus.

RESULTS

- In 2022, 2,790 patients (50%) with HHF+T2DM were identified as SOLOIST-like versus 2,830 patients not-SOLOIST-like according to clinical criteria.
- The SOLOIST-like group was younger with higher body mass index and had a higher prevalence of HFrEF compared to the not-SOLOIST-like group (62% vs 58%, p<0.01) (Table 2).
- SOLOIST-like patients were more often treated with mineralocorticoid receptor antagonist - MRA (38% vs 34%, p<0.01), as well as newer HF medications such as angiotensin receptor-neprilysin inhibitor - ARNI (9% vs 7%, p<0.01) or sodium-glucose cotransporter-2 inhibitors – SGLT2i (25% vs 22%, p<0.01) despite the overall low prescription rate.
- No significant difference was found in 1-year KM survival analysis among the two cohorts (HR 0.91; 95% CI 0.79-1.04,p=0.30) (Figure 2).

Table 2. Baseline characteristics and medication based on study groups.

Variable	SOLOIST-like N=2,790	Not-SOLOIST-like N=2,830	P values
Age, mean ± SD	68.6 ± 11.0	71.0 ± 12.3	<0.01
Female sex, %	44%	43%	0.38
Body mass index, kg/m^2	33.1 ± 7.5	31.8 ± 7.3	<0.01
Not Hispanic or Latino, %	85%	84%	0.32
Hispanic or Latino, %	14%	15%	
White, %	74%	73%	0.99
Black or African American, %	10%	10%	
Current smoker, %	26%	24%	0.09
Arterial hypertension, %	81%	80%	0.45
Hyperlipidemia, %	70%	70%	0.81
Coronary artery disease, %	61%	60%	0.28
Peripheral artery disease, %	16%	17%	0.40
Chronic kidney disease*, %	40%	38%	0.09
HFrEF, %	62%	58%	<0.01
HFpEF, %	38%	42%	<0.01
Beta-blockers, %	82%	83%	0.34
ACEi/ARB, %	89%	88%	0.52
MRA, %	38%	34%	<0.01
ARNI, %	9%	7%	<0.01
SGLT2i, %	25%	22%	<0.01

HHF: hospitalization for heart failure, T2DM: type 2 diabetes mellitus, HFpEF: heart failure with preserved ejection fraction, HFrEF: heart failure with reduced ejection fraction, ACEi: angiotensin converting enzyme inhibitor, ARB: angiotensin receptor blocker, ARNI: angiotensin receptor-neprilysin inhibitor, eGFR: estimated glomerular filtration rate, MRA: mineralocorticoid receptor antagonist, SGLT2i: sodium-glucose cotransporter 2 inhibitor. * defined as eGFR 30-59 mL/min/1.73m^2

Figure 2. 1-year Kaplan-Meier cumulative rates for death of study groups.

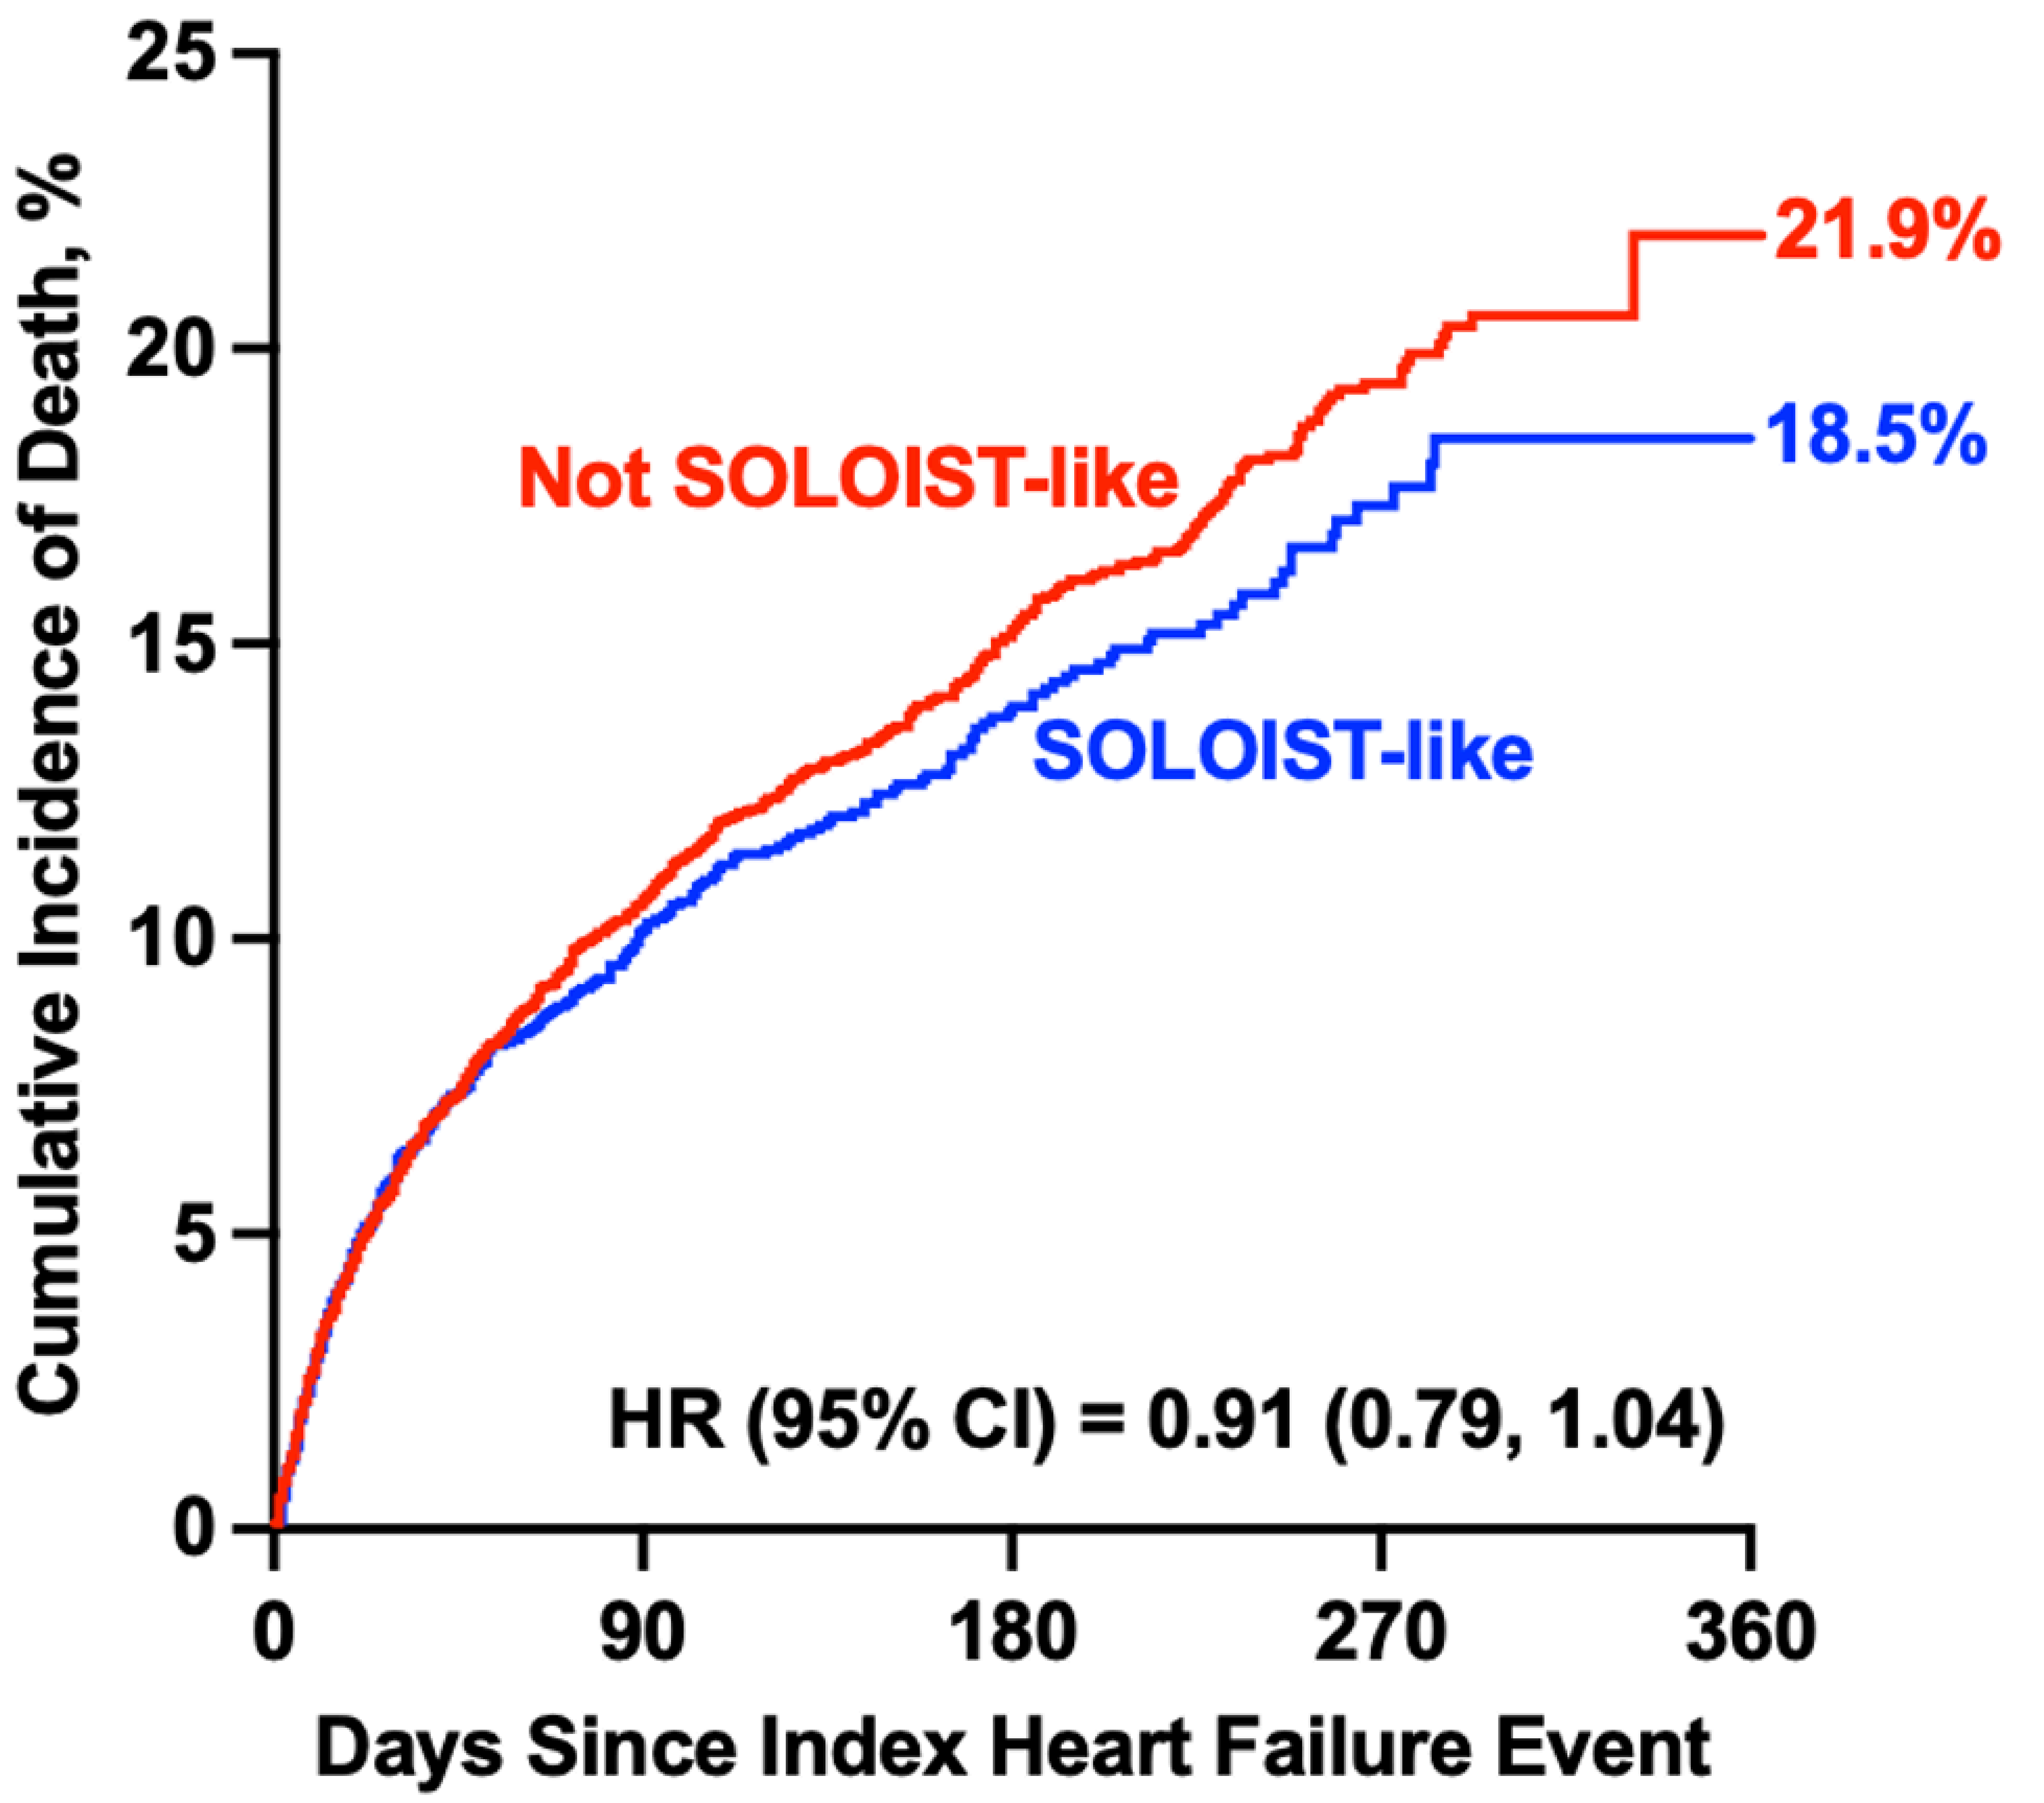
21.9%
18.5%
Not SOLOIST-like
SOLOIST-like
HR (95% CI) = 0.91 (0.79, 1.04)
Cumulative Incidence of Death, %
Days Since Index Heart Failure Event
0
90
180
270
360
0
5
10
15
20
25

CONCLUSIONS

Real-world data from a single US health system suggest that the SOLOIST-WHF trial results are very generalizable to the real-world population. Encouragingly, 25% of patients are being treated with SGLT2i in 2022, but efforts are needed to improve this and adherence to all guideline-directed medical therapy, including ARNI and MRA.

REFERENCES

1) Bhatt DL et al. Sotagliflozin in Patients with Diabetes and Recent Worsening Heart Failure. New England Journal of Medicine 2021;384(2):117–28.

2) Canonico ME et al. Uptake of Newer Guideline-Directed Therapies in Heart Failure Patients With Diabetes or Chronic Kidney Disease. JACC Heart Fail 2022;10(12):989–91.

FUNDING

This research received no external funding.

CONTACT INFORMATION

Mario Enrico Canonico, MD, PhD. CPC Clinical Research, Department of Medicine, University of Colorado, Aurora, CO, USA. email: marioenrico.canonico@cpcmed.org